Recombinant Alkaline Phosphatase modulates inflammation and injury in two rat models of AKI

Bülent Ergin[1], Asli Kandil[2], Ebru Gurel[2], Andrea van Elsas[3] and Can Ince[1]. 1) Department of Translational Physiology, Academic Medical Center, University of Amsterdam, Netherlands, 2) Department of Biology, University Istanbul, Turkey, and 3) AM Pharma, Bunnik, Netherlands.

Background

Understanding Alkaline Phosphatase Mode-of-Action in treating Acute Kidney Injury. Purified Bovine intestinal Alkaline Phosphatase (BiAP) demonstrated intriguing clinical activity as a potential therapeutic option for sepsis-associated acute kidney injury (Heemskerk et al. 2009, Crit.Care Med. 37: 417; Pickkers et al., 2012, Crit.Care 23:R14). The mechanism of action of Alkaline phosphatase in the treatment of acute kidney injury in sepsis patients remains to be determined. A novel human chimeric recombinant alkaline phosphatase (recAP) was constructed by swapping the stability (crown) domain from human intestinal AP with that derived from human placental AP.

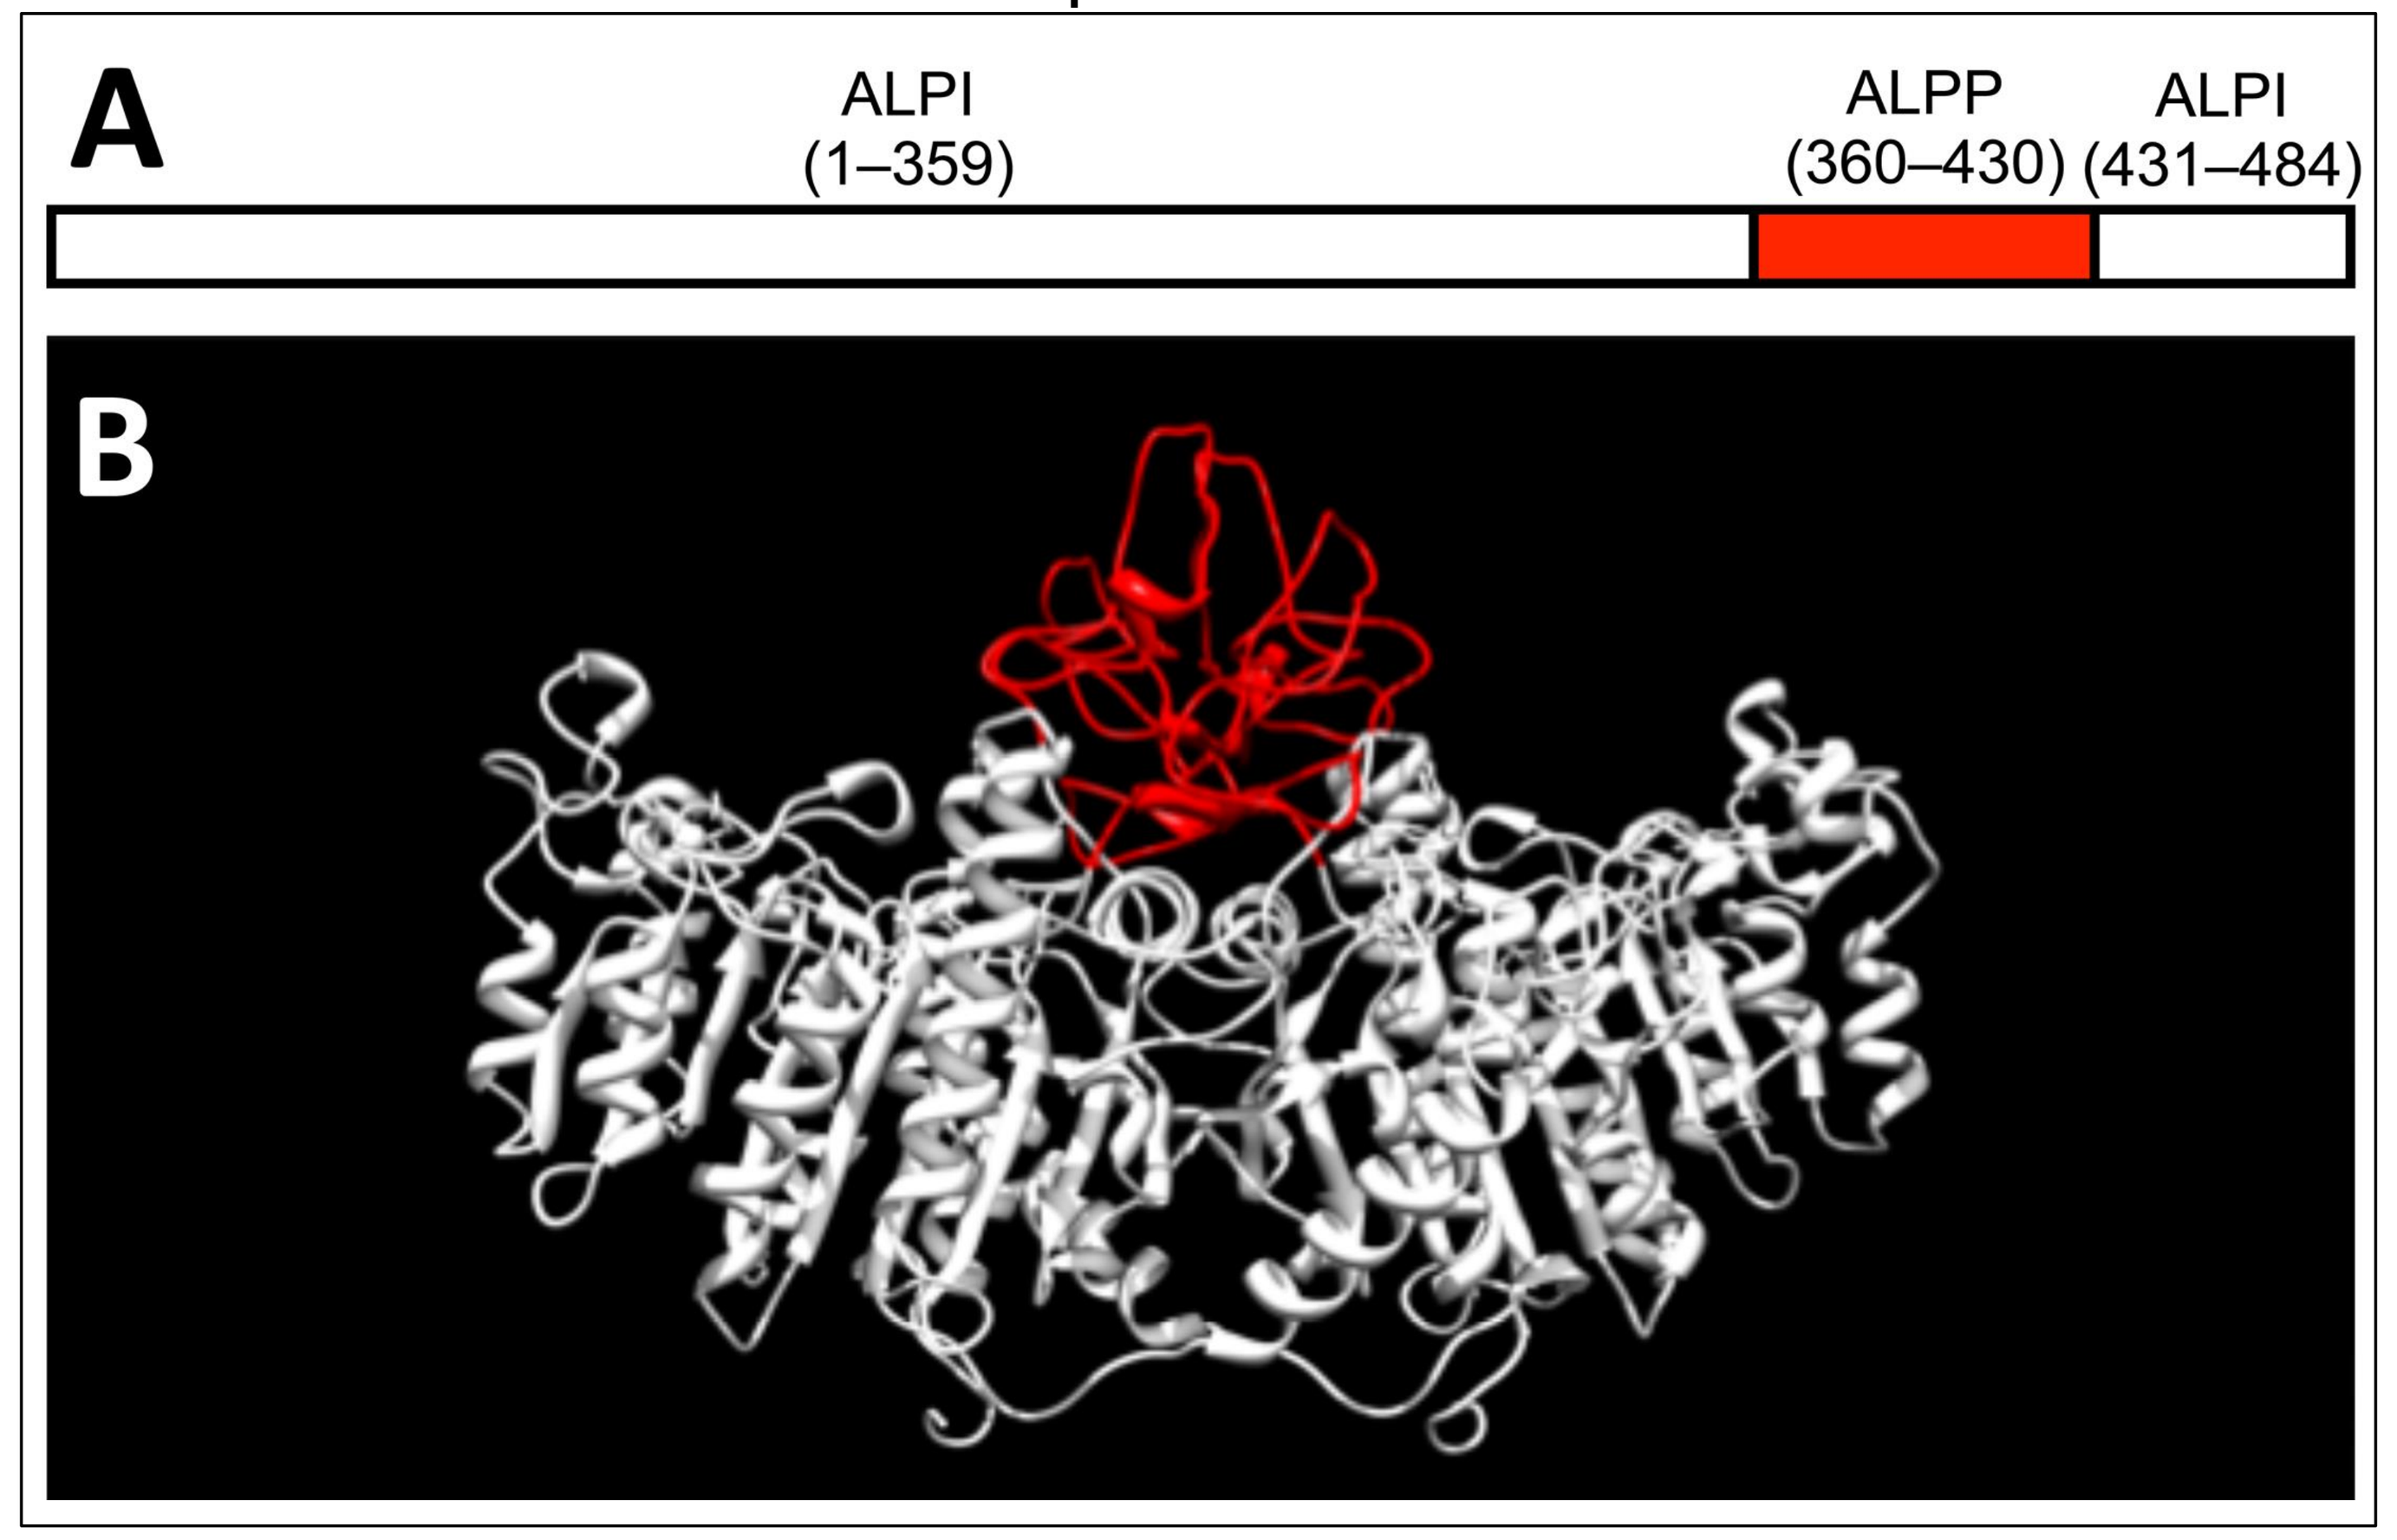
A
ALPI
(1–359)
ALPP
(360–430)
ALPI
(431–484)
B

Fig 1. *A novel recombinant human alkaline phosphatase, recAP, consists of the stable crown domain of human Placental AP (red), fused to the activity domain of human Intestinal AP. (Kiffer-Moreira et al., 2014)*

Compared to BiAP, recAP retains all enzyme activity but gains in stability and PK properties**.** To study the activity and mode-of-action of recAP, the protein was applied to rats during the first hours following induction of acute kidney by ischemia-reperfusion (I/R model) or LPS injection (Septic shock model; Legrand et al., 2011). Rats were kept warm and received fluid resuscitation as required for the duration of the experiments. Animals were instrumented for real-time assessment of hemodynamics. Kidneys were exposed to quantify oxygen delivery and expenditure. Blood and urine was collected for biochemical analysis and inflammatory kidney damage markers.

Abbreviations used

AKI - acute kidney injury; **BAX** – Bcl2 associated X protein; **IL-6** – interleukin 6; **iNOS** – inducible NO synthase; **I/R** - ischemia/reperfusion; **L-FABP** - L fatty acid binding protein; **LPS** - lipopolysaccharide; **MAP** - mean arterial pressure; **MDA** – malondialdehyde; **NGAL** – neutrophil gelatinase-associated lipocalin; **RBF** - renal blood flow; **recAP** – recombinant alkaline phosphatase; **RVR** - renal vascular resistance;

I/R-induced AKI

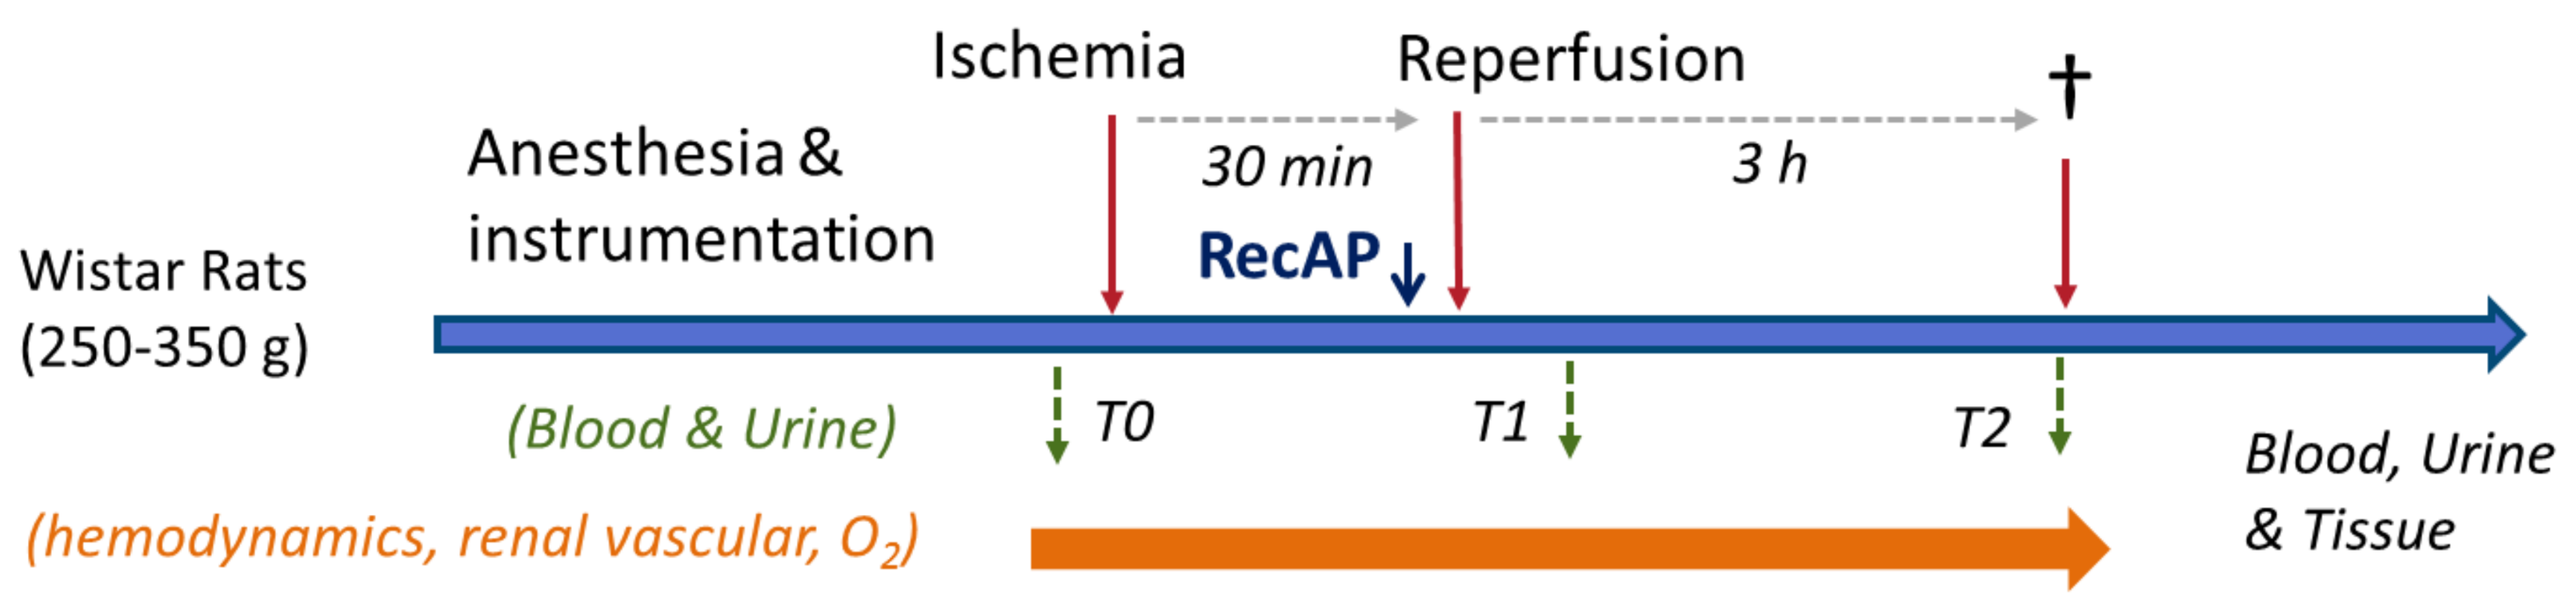
Wistar Rats
(250-350 g)
Anesthesia &
instrumentation
Ischemia
30 min
RecAP
Reperfusion
3 h
†
(Blood & Urine)
T0
T1
T2
(hemodynamics, renal vascular, O2)
Blood, Urine
& Tissue

Fig 2. *Schematic set-up of the I/R induced rat AKI experiment. Within the first few hours post-induction real-time data on hemodynamics and oxygen consumption were collected and tissue harvested for analysis of markers of inflammation and kidney damage.*

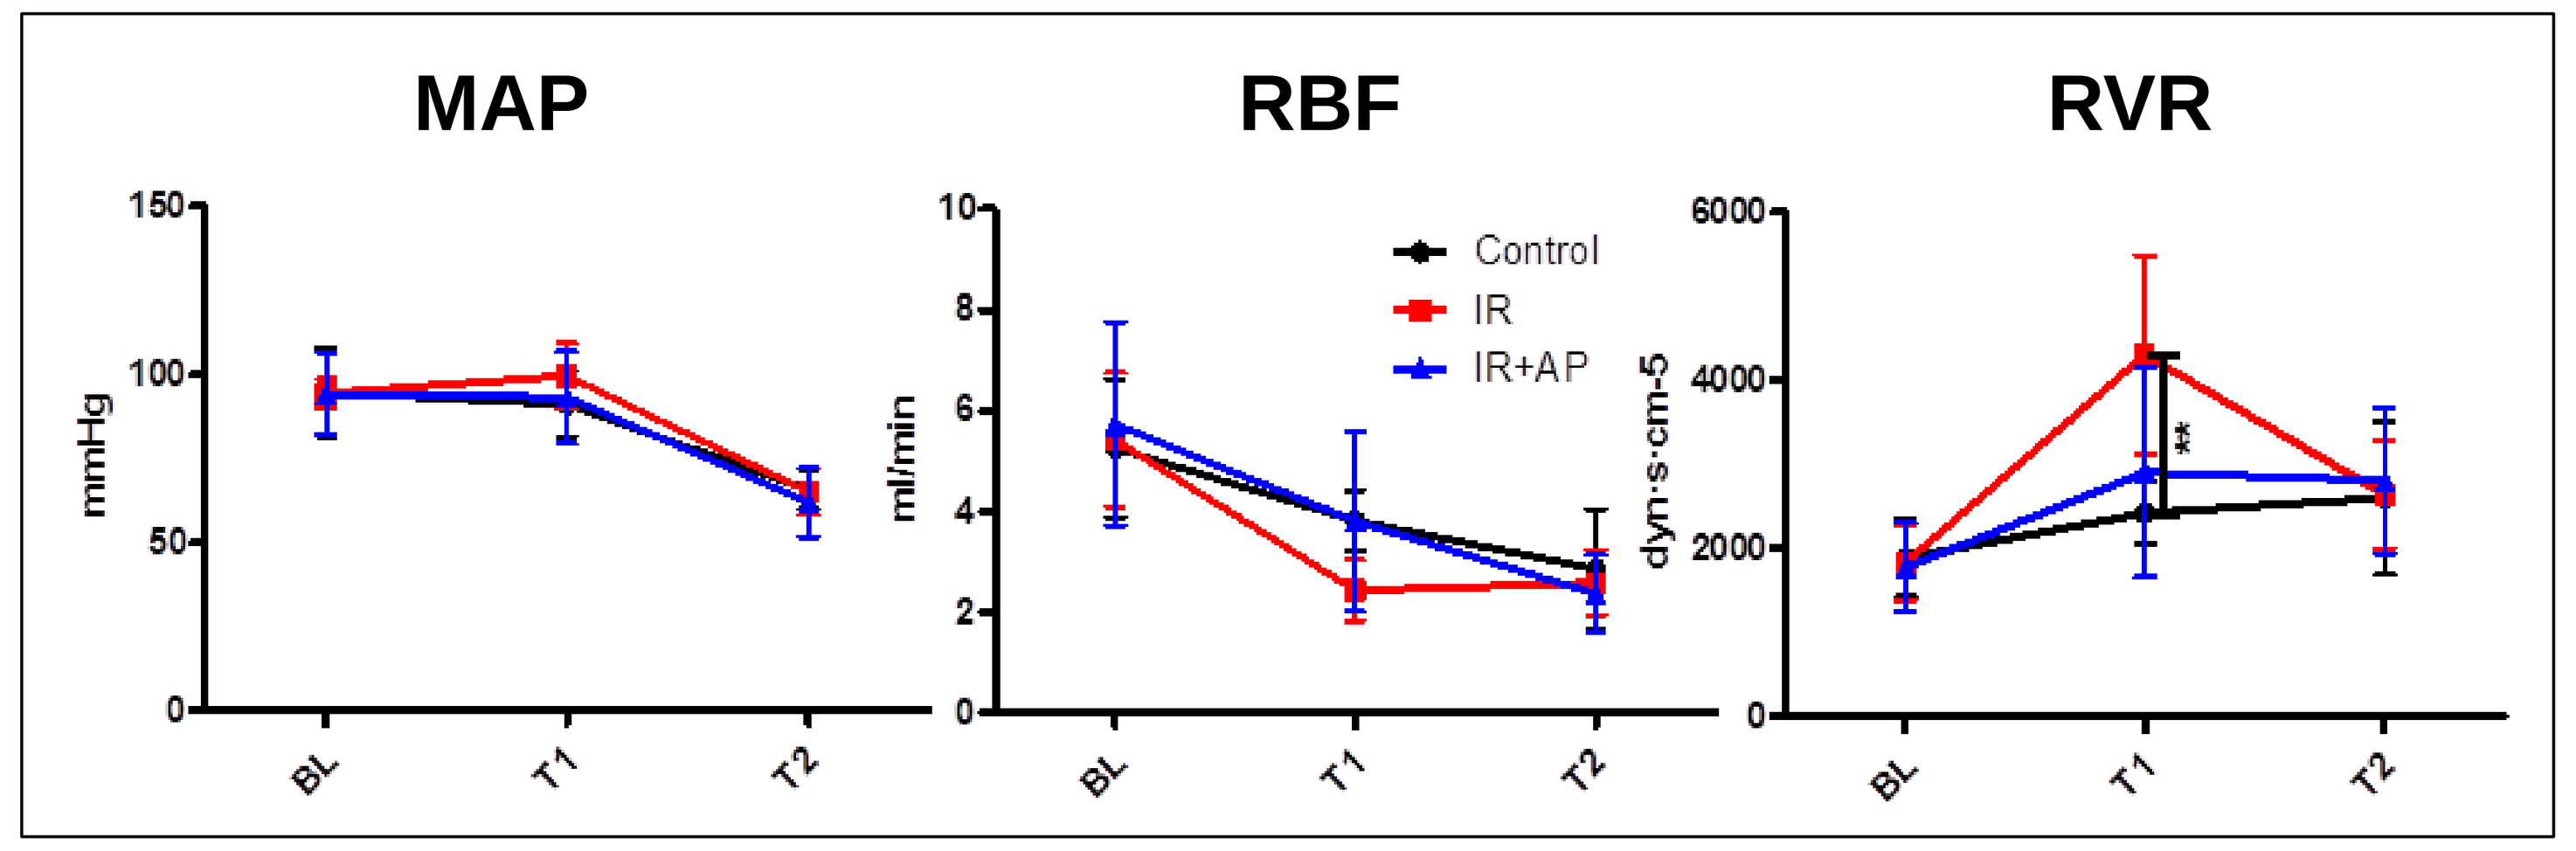
MAP
RBF
RVR
mmHg
ml/min
dyn·s·cm-5
Control
IR
IR+AP
BL
T1
T2

Fig 3. *Hemodynamic response to I/R and treatment with recAP. Semi-therapeutic administration of recAP demonstrated little effect on systemic hemodynamics (MAP) , but significantly affected renal vascular resistance immediately post-reperfusion.*

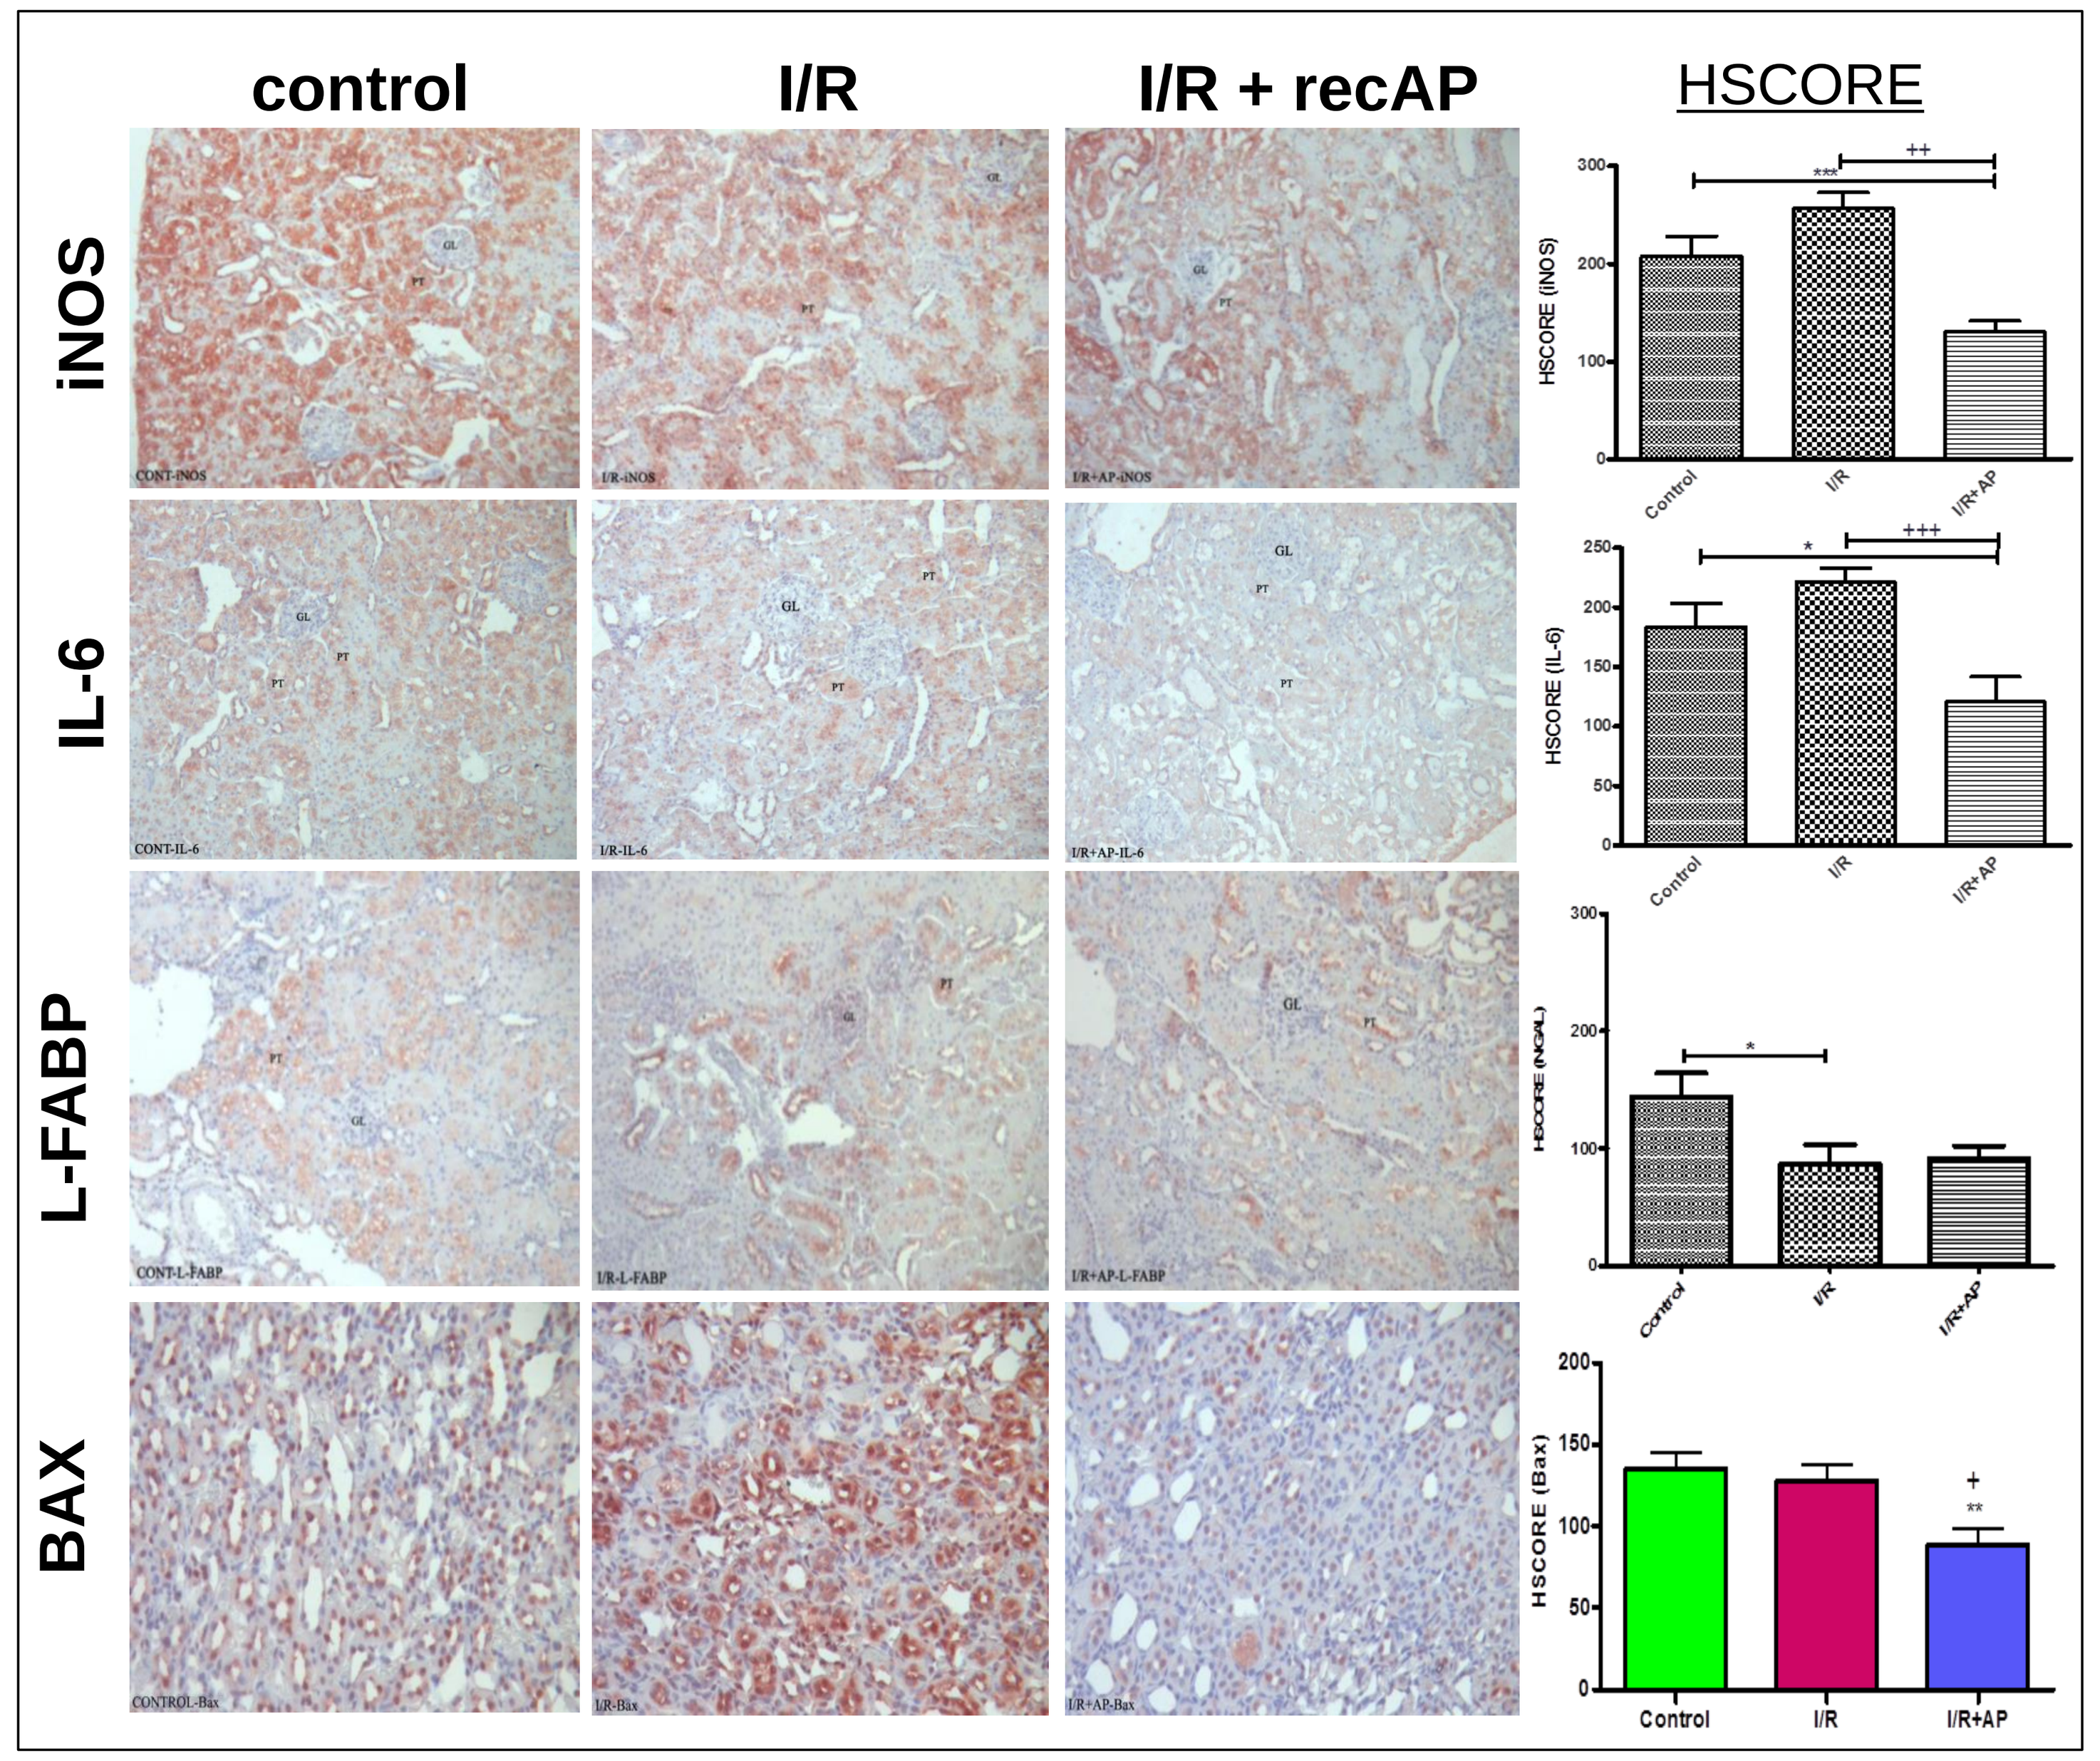
control
I/R
I/R + recAP
HSCORE
iNOS
IL-6
L-FABP
BAX
HSCORE (iNOS)
HSCORE (IL-6)
HSCORE (NGAL)
HSCORE (Bax)
Control
I/R
I/R+AP

Fig 4. *Kidneys harvested 3 hours post-treatment were stained for various markers of inflammation (iNOS, IL-6) and renal injury response (L-FABP, Bax). Staining was quantified using histological scoring methodology (HSCORE). RecAP inhibited iNOS and IL-6. Damage markers were not induced by I/R and not regulated by recAP.*

LPS-induced AKI

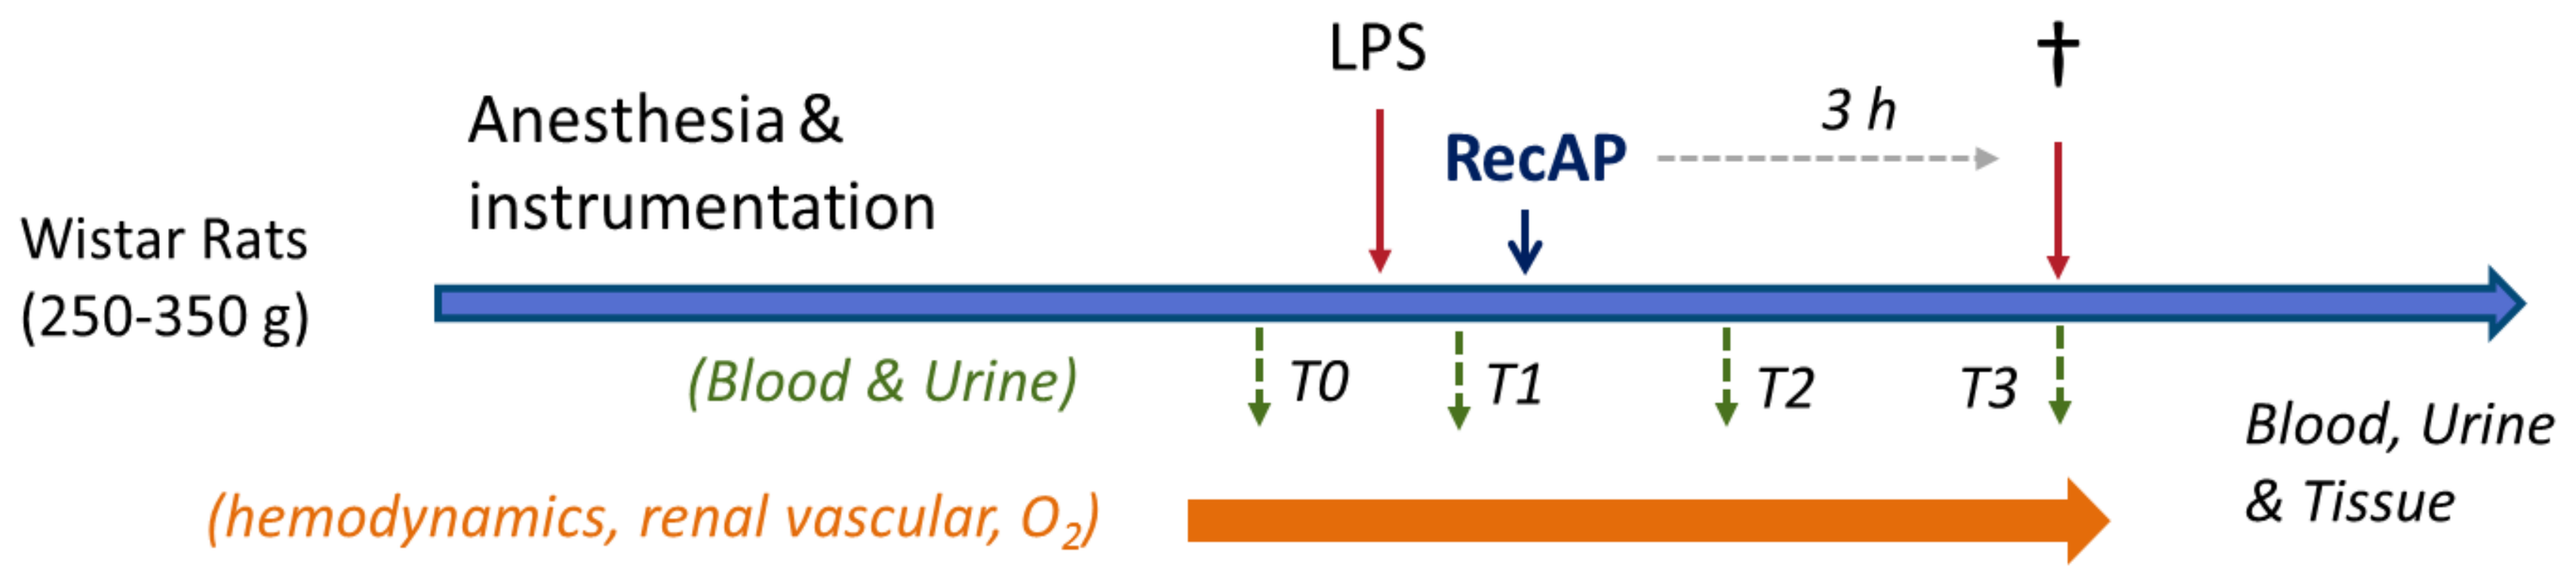
Wistar Rats
(250-350 g)
Anesthesia &
instrumentation
LPS
RecAP
3 h
†
(Blood & Urine)
T0
T1
T2
T3
(hemodynamics, renal vascular, O2)
Blood, Urine
& Tissue

Fig 5. *Schematic set-up of the LPS-induced rat AKI experiment. Within the first few hours post-induction real-time data on hemodynamics and oxygen consumption were collected and tissue harvested for analysis of markers of inflammation and kidney damage.*

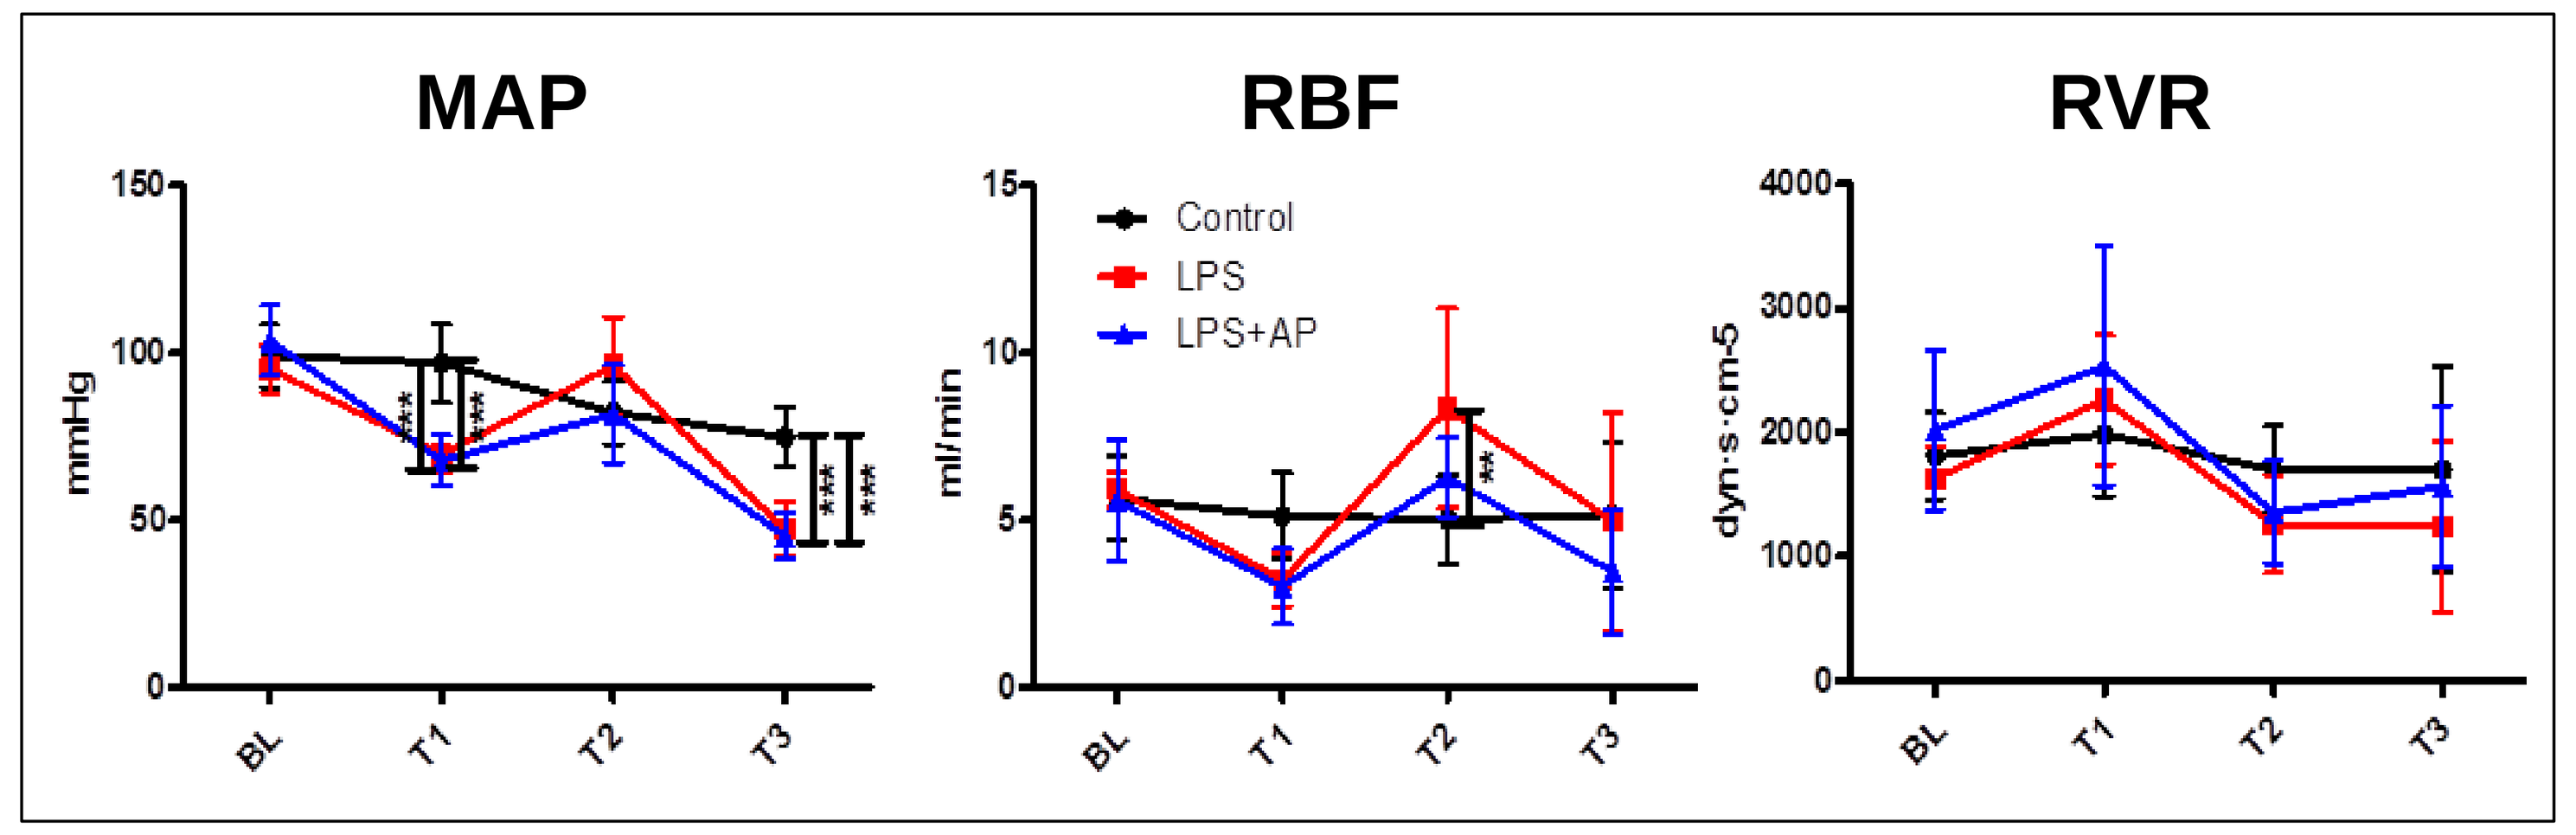
MAP
RBF
RVR
mmHg
ml/min
dyn·s·cm-5
Control
LPS
LPS+AP
BL
T1
T2
T3

Fig 6. *Hemodynamic response to LPS and treatment with recAP. Semi-therapeutic administration of recAP demonstrated little effect on systemic hemodynamics (MAP) , but significantly affected renal blood flow.*

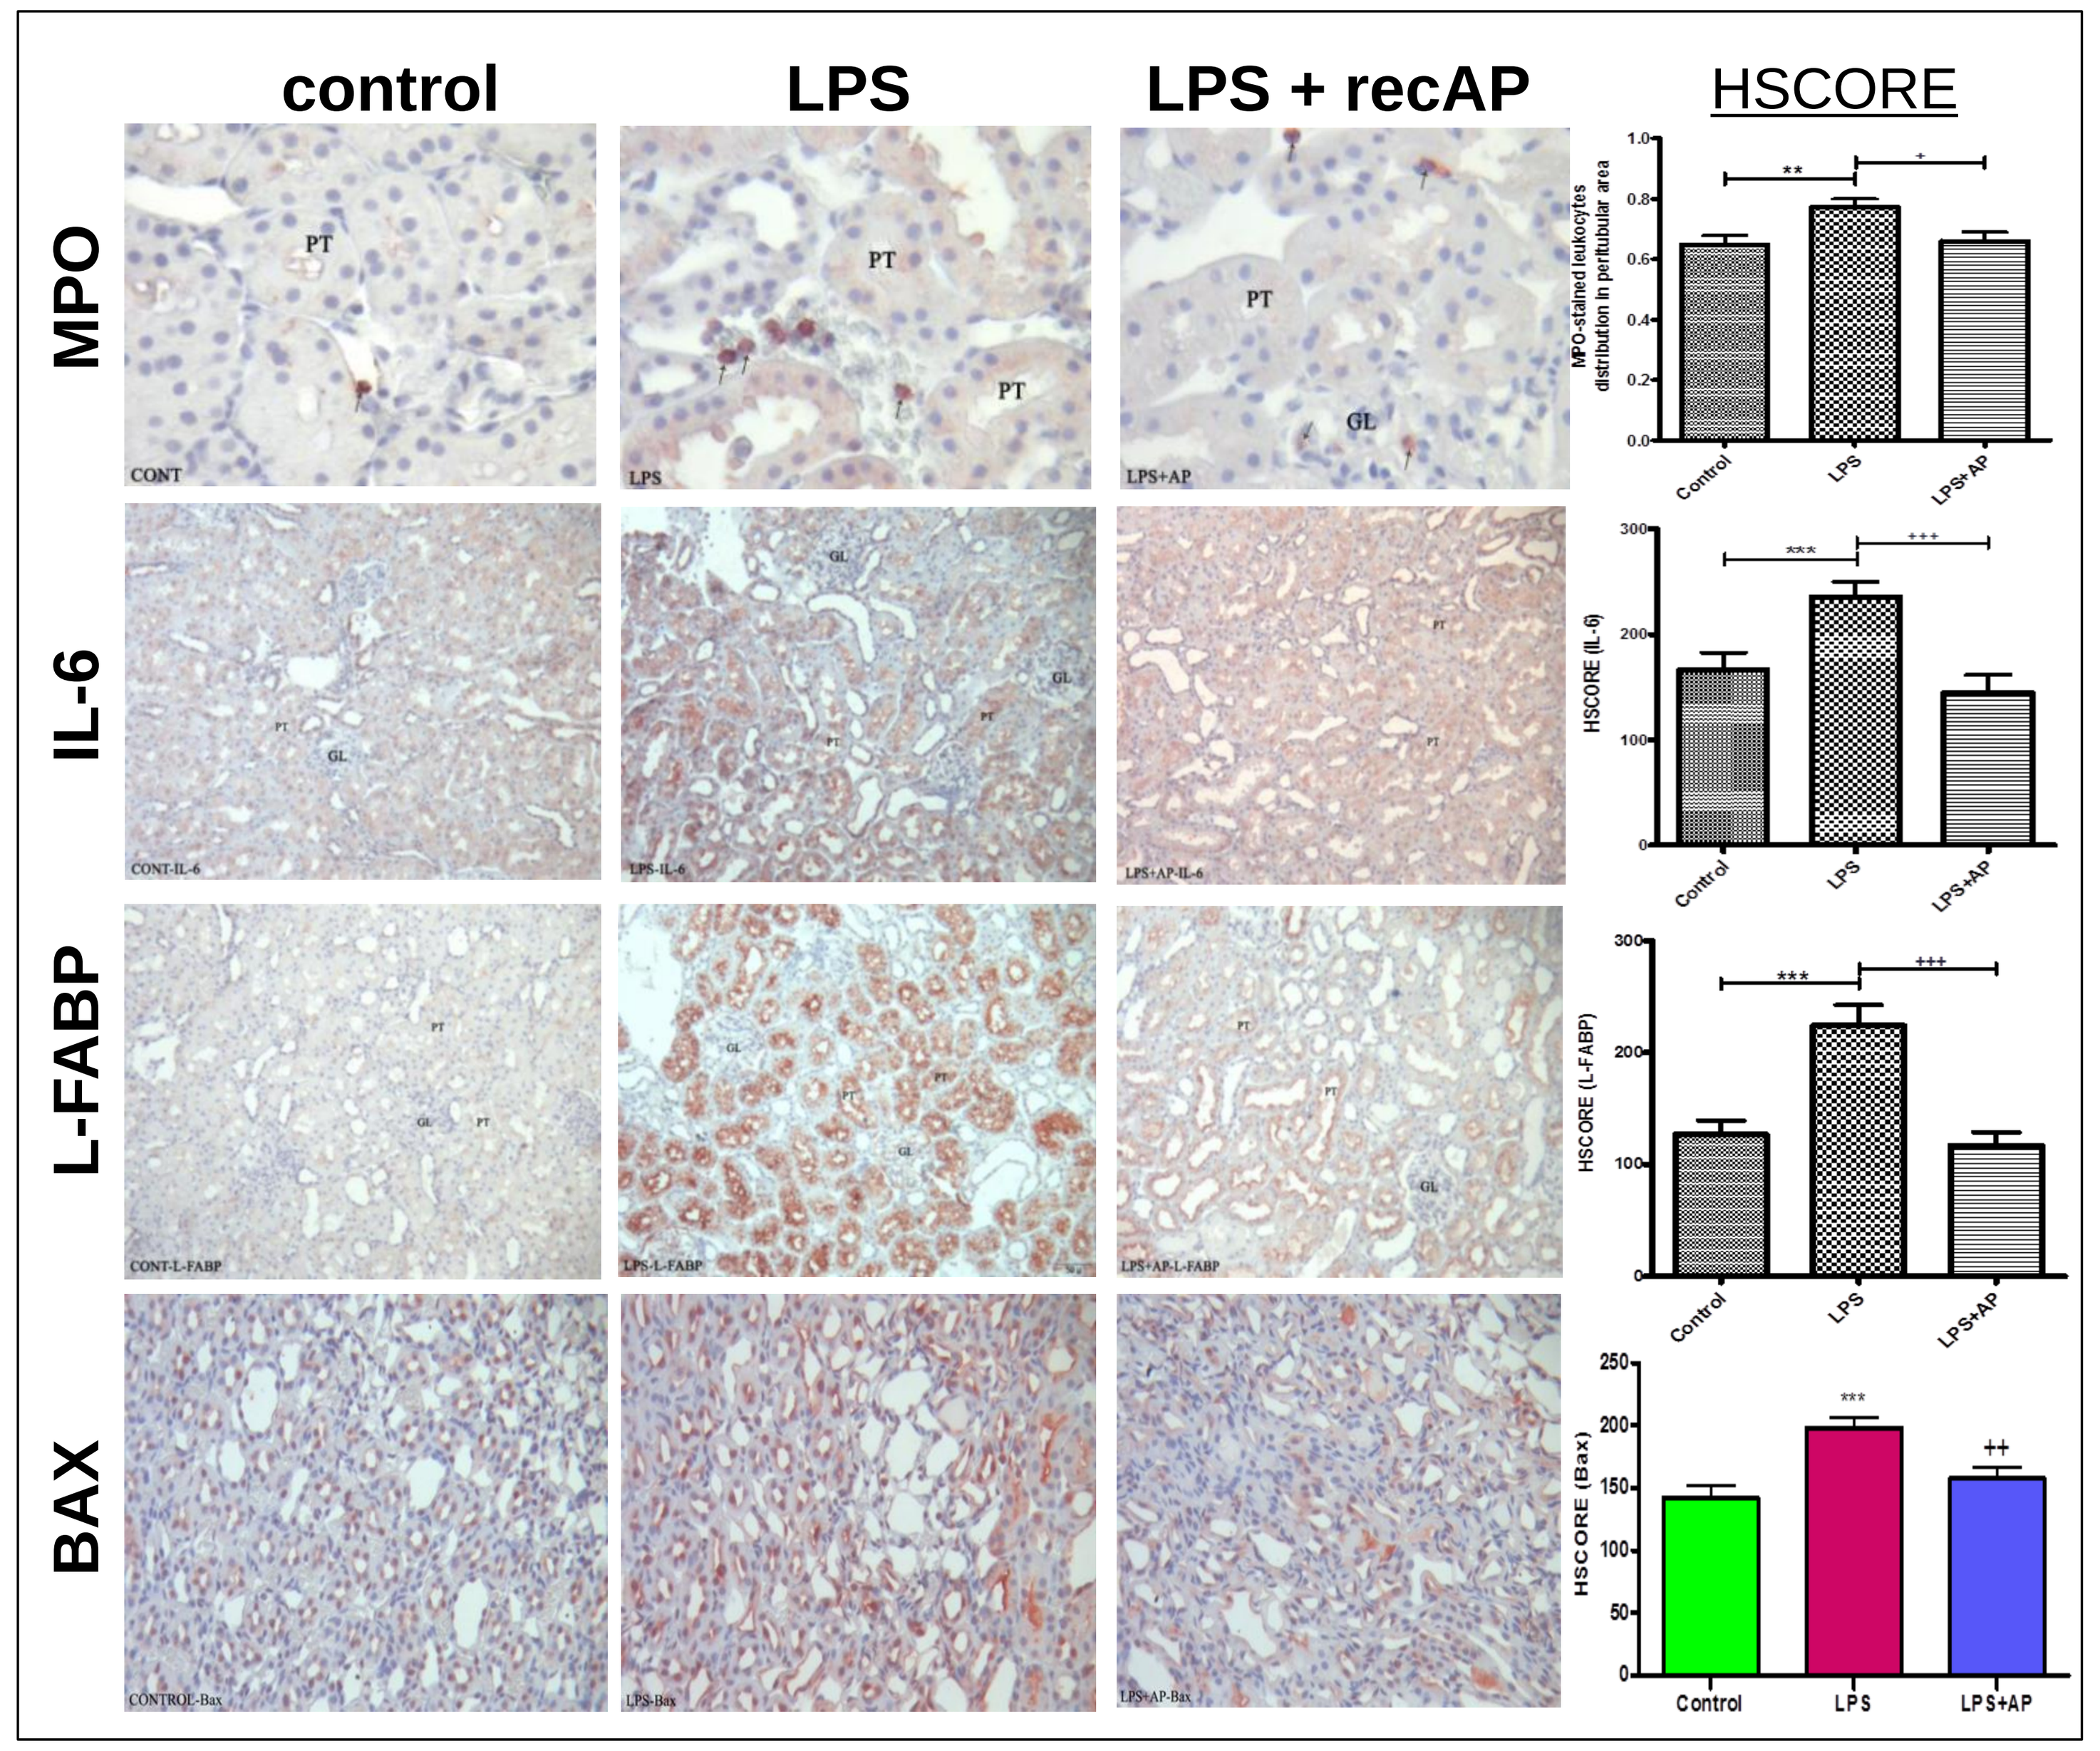
control
LPS
LPS + recAP
HSCORE
MPO
IL-6
L-FABP
BAX
MPO-stained leukocytes distribution in peritubular area
HSCORE (IL-6)
HSCORE (L-FABP)
HSCORE (Bax)
Control
LPS
LPS+AP

Fig 7. *Kidneys harvested 3 hours post-treatment were stained for various markers of inflammation (MPO, IL-6) and renal injury response (L-FABP, Bax). Similar data were recorded for iNOS and NGAL.RecAP completely inhibited LPS mediated induction of neutrophil infiltration (MPO), inflammatio (iNOS, IL-6), and injury (L-FABP, NGAL, Bax).*

Conclusions

In rats, mild (I/R) or severe (LPS) acute kidney injury was induced and kidney disease followed for up to 3 h post-induction. A single i.v. dose of 1000 U/kg recAP modulated markers of:

- Renal vascular response (RBF, RVR)
- Leukocyte infiltration and inflammation (MPO, iNOS, IL-6)
- Renal injury and apoptosis (L-FABP, NGAL, Bax)

In the context of disease severity induced, and within the 3 h time-frame of the experiment, oxygenation (cortical, medullar O_2 delivery and consumption) and tubular function (Creatinine Clearance, FE_{Na2}) were not modulated.

Therefore it was concluded that in these animals RecAP inhibited acute renal inflammation and markers of tissue damage

Hypothetical Mode-of-Action

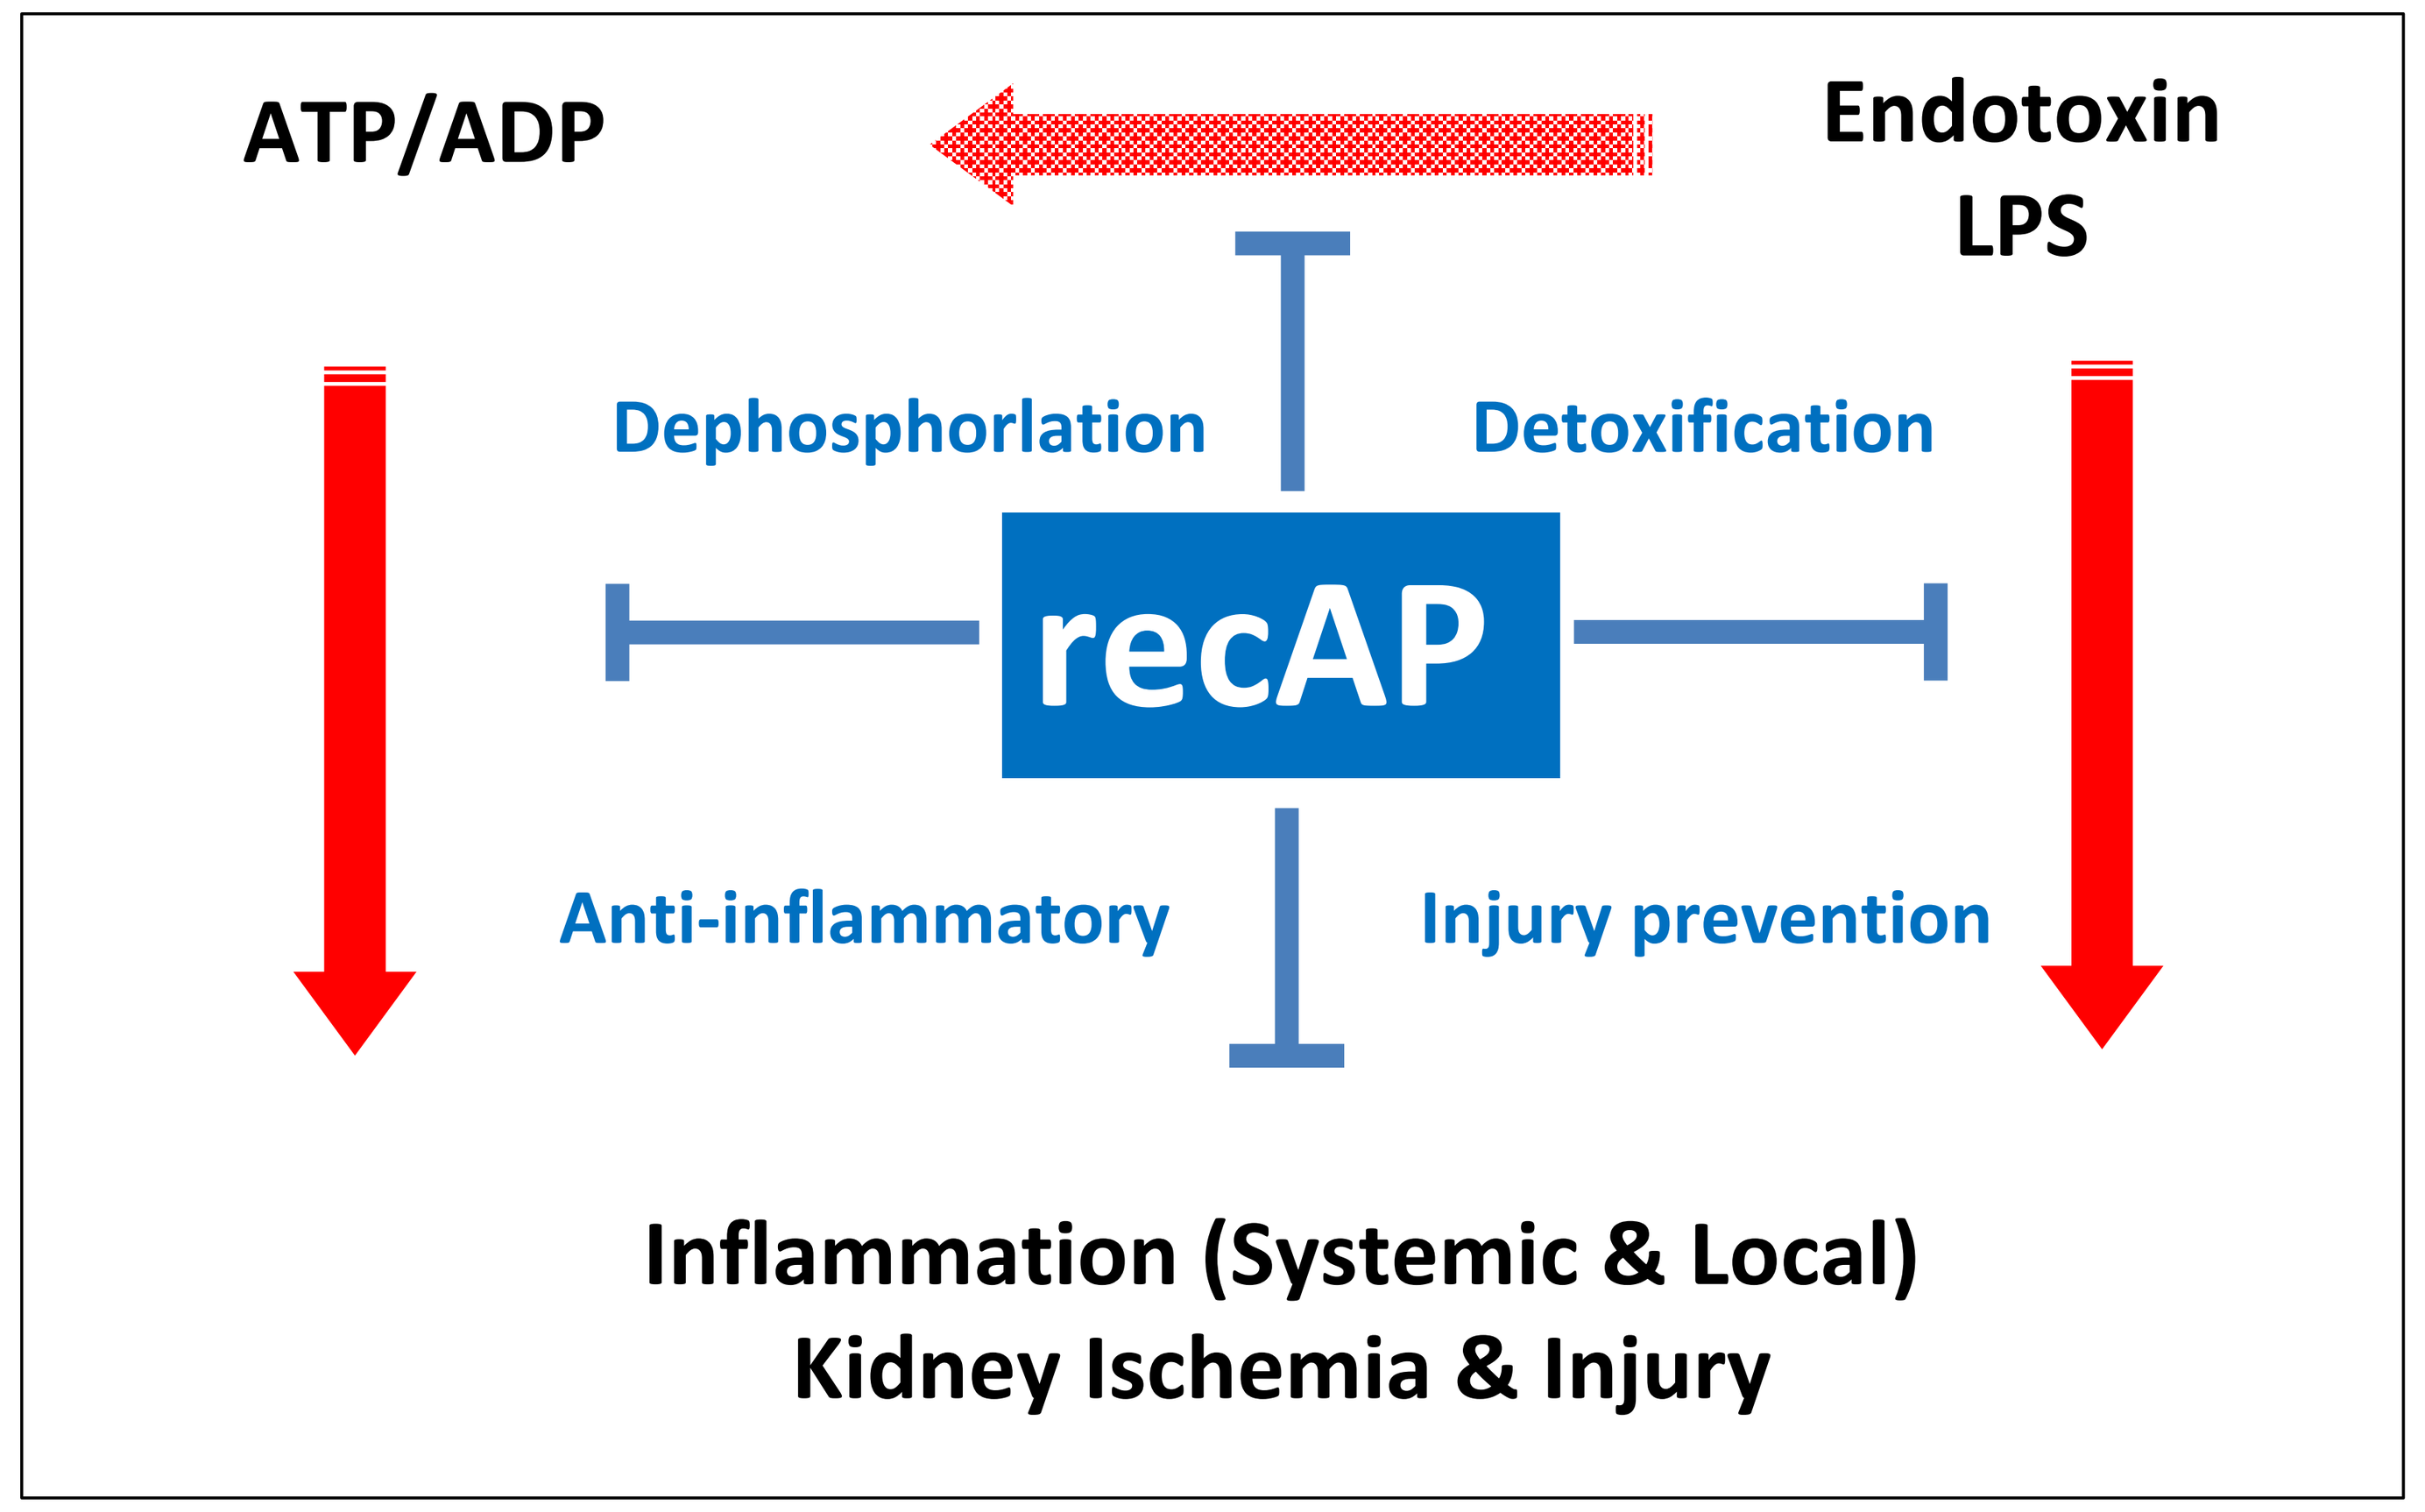
ATP/ADP
Endotoxin
LPS
Dephosphorlation
Detoxification
recAP
Anti-inflammatory
Injury prevention
Inflammation (Systemic & Local)
Kidney Ischemia & Injury

Fig 8. *RecAP dephosphorylates potent immunostimulatory triggers, such as ATP, ADP released into the extracellular space in the kidney under stress and LPS present in the circulation and the kidney. As such, recAP suppresses inflammation and renal injury and enables repair of kidney function (Peters et al., 2013).*

References

1. *Heemskerk S, et al. (APSEP Study Group). Alkaline phosphatase treatment improves renal function in severe sepsis or septic shock patients. Crit Care Med. 2009 Feb;37(2):417-23*
2. *Kiffer-Moreira T, et al. Catalytic signature of a heat-stable, chimeric human alkaline phosphatase with therapeutic potential. PLOS One 2014, in press.*
3. *Legrand M, et al. The role of renal hypoperfusion in development of renal microcirculatory dysfunction in endotoxemic rats. Intensive Care Med. 2011 Sep;37(9):1534-42*
4. *Peters E, et al. Alkaline phosphatase as a treatment of sepsis-associated acute kidney injury. J Pharmacol Exp Ther. 2013 Jan;344(1):2-7*
5. *Pickkers et al. Alkaline phosphatase for treatment of sepsis-induced acute kidney injury: a prospective randomized double-blind placebo-controlled trial. Crit Care. 2012 Jan 23;16(1):R14*